Discovery and Preclinical Characterization of INCB018424, a Selective JAK2 Inhibitor for the Treatment of Myeloproliferative Disorders

Jordan Fridman*, Roberto Nussenzveig^, Phillip Liu*, Stacey Shepard*, James Rodgers*, Timothy Burn*, Patrick Haley*, Peggy Scherle*, Robert Newton*, Gregory Hollis*, Steven Friedman*, Srdan Verstovsek ^, and Kris Vaddi*

*Incyte Corporation, Wilmington, DE, and 2Leukemia Department, ^M.D. Anderson Cancer Center, Houston, TX

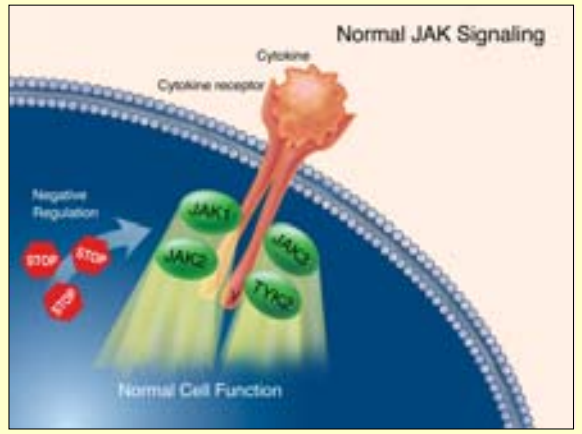

Normal JAK Signaling
Cytokine
Cytokine receptor
Negative Regulation
STOP
JAK1
JAK2
JAK3
TYK2
Normal Cell Function

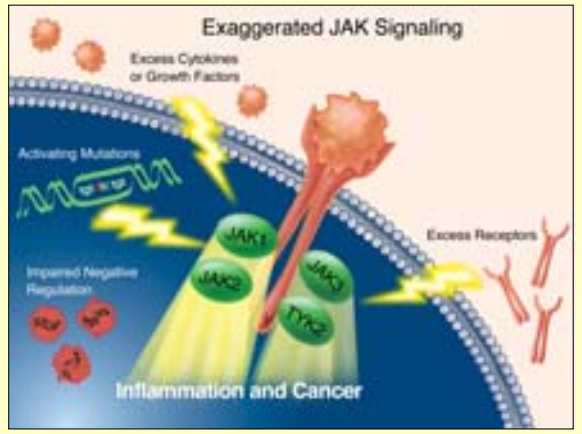

Exaggerated JAK Signaling
Excess Cytokines or Growth Factors
Activating Mutations
Impaired Negative Regulation
Excess Receptors
JAK1
JAK2
JAK3
TYK2
Inflammation and Cancer

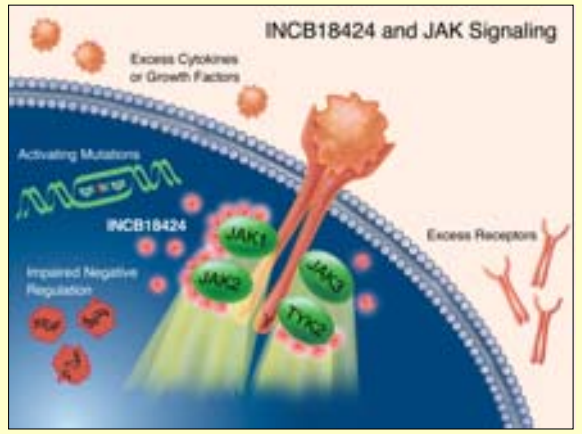

INCB18424 and JAK Signaling
Excess Cytokines or Growth Factors
Activating Mutations
INCB18424
Impaired Negative Regulation
Excess Receptors
JAK1
JAK2
JAK3
TYK2

ABSTRACT

Activating mutations in Janus kinase 2 (JAK2) have recently been identified in the majority of Philadelphia chromosome negative (Ph-) myeloproliferative disorders (MPDs). Importantly, constitutive JAK2 activation is oncogenic and, in murine models, recapitulates much of the pathobiology observed in MPD patients, suggesting that JAK2 inhibition may be of therapeutic benefit. Here we describe the identification and preclinical characterization of INCB018424, a potent, selective, and orally bioavailable inhibitor of the JAK2 now in clinical trials. INCB018424 was identified through an extensive medicinal chemistry effort designed to optimize potency, selectivity, pharmaceutical and pharmacokinetic properties. INCB018424 inhibits JAK2 at <1 nM and demonstrated >500-fold selectivity against a broad sampling of the kinome. The potency and selectivity of INCB018424 translated to exceptional cellular activity where it inhibited the proliferation of FDCP cells and BaF/3 cells expressing JAK2V617F with an IC50 of 100-130 nM, but not the proliferation of cell lines expressing activating mutations in either BCR-Abl or cKit (IC50 > 25 and 4 μM, respectively). The effect of INCB18424 on cell proliferation correlated well with reduced levels of phosphorylated JAK2 and STAT5 in the BaF/3 cell model, suggesting that the effect is mediated by pharmacological inhibition of JAK-STAT pathway. Interestingly, the activation of endogenous wild-type JAKs – by the addition of IL-3 – shifted the potency of INCB018424 in the BaF/3 model greater than five fold suggesting that cells expressing the mutated form of JAK2 may be more sensitive to INCB018424. Indeed, using cells harvested from patients with JAK2V617F-positive polycythemia vera (PV) in colony forming assays, we observed that INCB018424 inhibited the cytokine-independent formation of erythroid progenitor colonies (n=3) with an IC50 of 67nM while normal colony formation from healthy donors (n=3) was inhibited 50% at > 400 nM. Further, INCB018424 inhibited proliferation of PV patient samples (n=3) following *ex vivo* expansion of erythroid progenitors in serum free media, with an IC50 of 60 nM, similar to that observed in semi-solid media. In a mouse model of MPD, where implantation of BaF/3 cells expressing JAK2V617F results in rapid organomegaly and reduced survival, oral administration of INCB018424 was well tolerated and markedly reduced the splenomegaly. Using this animal model, we also demonstrated that selective JAK inhibition eliminates neoplastic cells from the spleen, liver, and bone marrow normalizing the histology of affected organs and significantly prolongs survival. As such, potent and selective JAK inhibitors such as INCB018424 hold great promise for the treatment of MPDs and other disease states associated with elevated JAK activity – a concept currently being tested clinically.

Disclosure: All contributors, excluding Drs. Nussenzveig and Verstovsek, are employees of Incyte Corporation.

1. Identification of potent (A) and selective (B) inhibitors of JAK2

A

Kinase Potency* (nM)

Compound	JAK1	JAK2	JAK3	Tyk2
INCB016562	7.5	2.9	1115	27
INCB018424	2.7	4.5	322	19

*performed at ~ cellular [ATP], 1mM

B

% Inhibition

Kinase	INCB016562	INCB018424
Abl	50	-1
Akt1	0	-5
AurA	46	5
AurB	ND	0
CDC2/CDK1	0	-3
CDK2	3	-2
CDK4	ND	3
CHK2	ND	-2
c-kit	ND	4
c-Met	ND	0
EGFR	9	0
EphB4	9	-1
ERK1	ND	0
ERK2	0	-4
FLT-1	8	5
HER2/ErbB2	ND	1
IGF1R	0	-2
FLT3	0	ND

% Inhibition

Kinase	INCB016562	INCB018424
IKKalpha	ND	-5
IKKbeta	0	29
JAK2	ND	100
JAK3	ND	95
JNK 1	ND	8
Lck	56	8
MEK1	0	-8
p38alpha	ND	-5
p70S6K	0	7
PKA	0	8
PKCalpha	0	9
Src	62	2
ZAP70	0	1
c-Raf	5	ND
Syk	27	ND
PDGFRb	0	ND
Axl	0	ND
Screening Conc.	100 nM	200 nM

2. INCB018424 retains it potency (A and B) and selectivity (A and C) in cell lines: no impact on Abl or cKIT driven proliferation

A

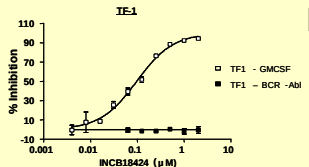

TF-1
% Inhibition
TF1 - GMCSF
TF1 – BCR -Abl
INCB18424 (μM)

	GM-CSF (JAK)	Bcr-Abl
IC_{50} (nM)	80 ± 30	> 25,000

B

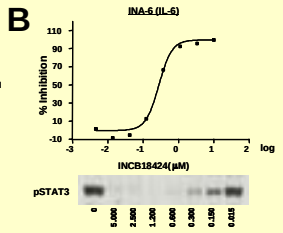

INA-6 (IL-6)
% Inhibition
INCB18424(μM)
log
pSTAT3

C

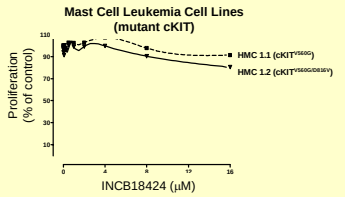

Mast Cell Leukemia Cell Lines
(mutant cKIT)
Proliferation (% of control)
HMC 1.1 (cKIT V560G)
HMC 1.2 (cKIT V560G/D816V)
INCB18424 (μM)

3. Oral administration of selective JAK inhibitors retards JAK mediated cytokine dependent tumor growth and reduces phosphorylation of STAT3

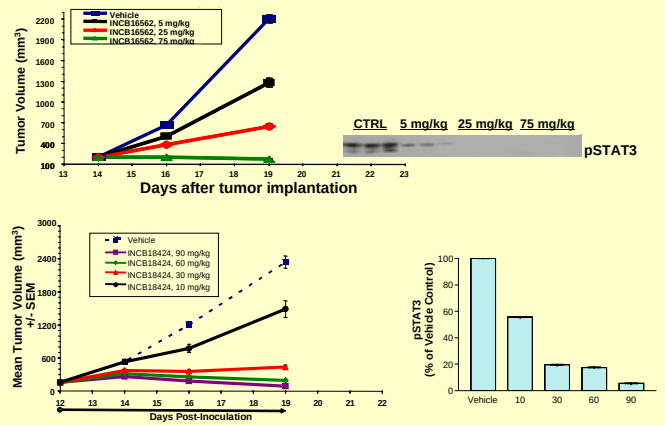

Tumor Volume (mm³)
Vehicle
INCB16562, 5 mg/kg
INCB16562, 25 mg/kg
INCB16562, 75 mg/kg
Days after tumor implantation
CTRL
5 mg/kg
25 mg/kg
75 mg/kg
pSTAT3
Mean Tumor Volume (mm³) +/- SEM
Vehicle
INCB18424, 90 mg/kg
INCB18424, 60 mg/kg
INCB18424, 30 mg/kg
INCB18424, 10 mg/kg
Days Post-Inoculation
pSTAT3 (% of Vehicle Control)
Vehicle
10
30
60
90

4. INCB018424 inhibits JAK/STAT signaling (A) and proliferation (B) in human MPD cell lines and primary patient cells (C)

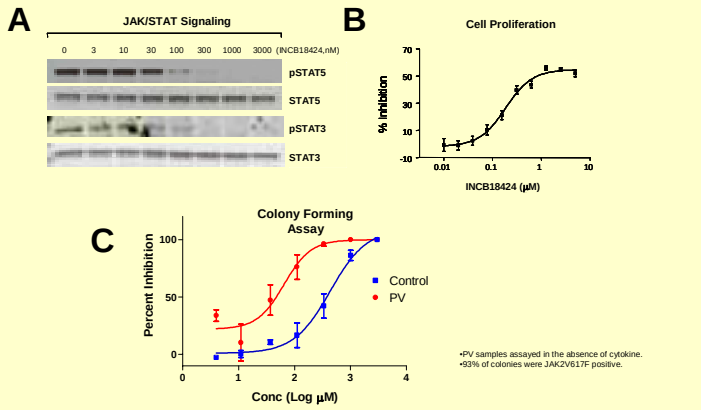

A
JAK/STAT Signaling
0
3
10
30
100
300
1000
3000 (INCB18424,nM)
pSTAT5
STAT5
pSTAT3
STAT3
B
Cell Proliferation
% Inhibition
INCB18424 (μM)
C
Colony Forming Assay
Percent Inhibition
Control
PV
Conc (Log μM)
•PV samples assayed in the absence of cytokine.
•93% of colonies were JAK2V617F positive.

Similar potency observed in expanded PV erythroid progenitor proliferation assays

5. INCB018424 diminishes proliferation (A) and JAK/STAT signaling (B) in the BaF/3JAK3V617F model resulting in induction of apoptosis (C)

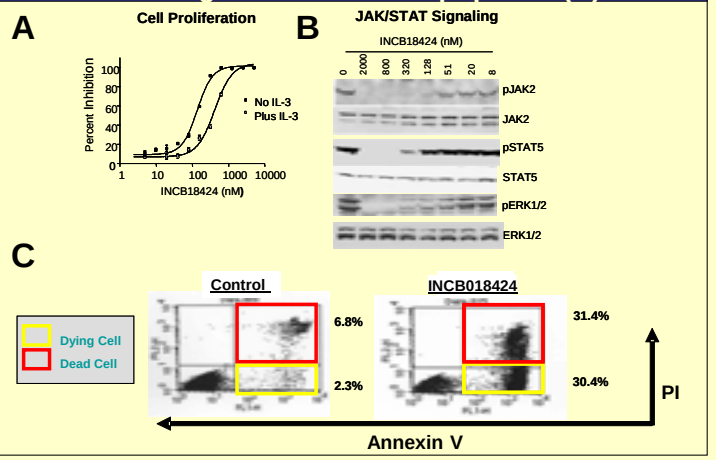

A
Cell Proliferation
Percent Inhibition
No IL-3
Plus IL-3
INCB18424 (nM)
B
JAK/STAT Signaling
INCB18424 (nM)
0
2000
800
320
128
51
20
8
pJAK2
JAK2
pSTAT5
STAT5
pERK1/2
ERK1/2
C
Dying Cell
Dead Cell
Control
INCB018424
6.8%
2.3%
31.4%
30.4%
PI
Annexin V

6. Selective JAK inhibitors improve survival (A) and markedly reduce splenomegaly (B) in a model of JAK2V617F malignancy

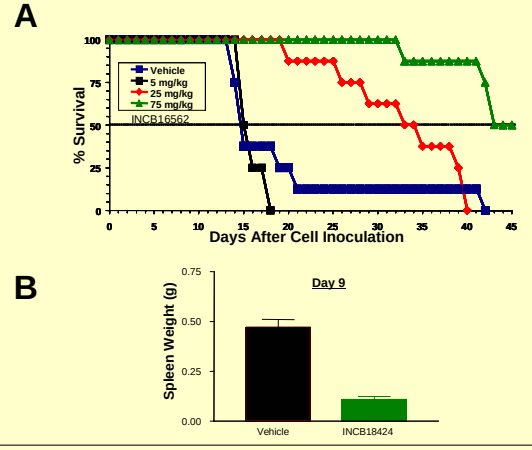

A
% Survival
Vehicle
5 mg/kg
25 mg/kg
75 mg/kg
INCB16562
Days After Cell Inoculation
B
Day 9
Spleen Weight (g)
Vehicle
INCB18424

7. Histological identification of neoplastic cell elimination from spleen (A), liver (B) and bone marrow (C) by a selective JAK inhibitor

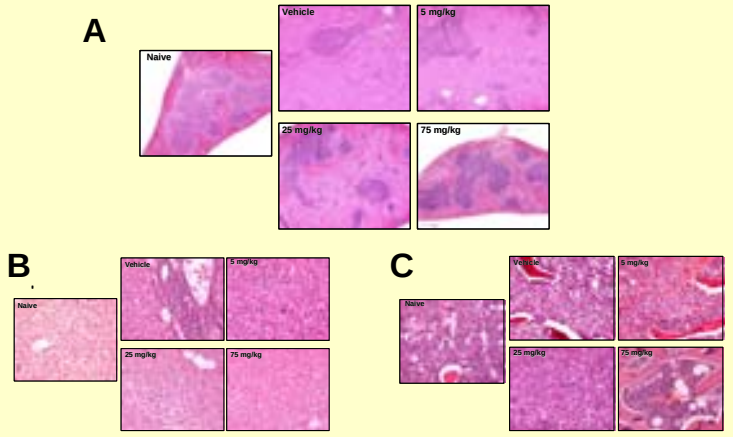

A
Naive
Vehicle
5 mg/kg
25 mg/kg
75 mg/kg
B
C

CONCLUSIONS

- Incyte has discovered a number of potent and selective JAK inhibitors
- INCB018424 is a potent and selective JAK2 inhibitor
- Selective JAK inhibition reduces cytokine-dependent JAK/STAT signaling and tumor growth *in vivo*
- In cells expressing JAK2V617F, INCB018424 reduces JAK/STAT signaling, proliferation, and colony formation and induces apoptosis
- Oral administration of selective JAK inhibitors improves survival and reduces splenomegaly in an *in vivo* model of JAK2V617F driven malignancy
- INCB018424 has demonstrated promising clinical activity in patients with myelofibrosis (Abstract #558)

Incyte

The Drive to Discover. The Experience to Deliver.